Early and Sustained Clinically Meaningful Gross Motor Improvements in a Phase 1/2 Study of NGN-401, a Novel Regulated AAV9 Gene Therapy, in Rett Syndrome

Poster #265

Jenny Downs[1], Melissa Licari[1], Bernhard Suter[2], Tim A. Benke[3], David Lieberman[4], Elizabeth Berry-Kravis[5], Aleksandra Jacobs[6], Carolyn Ellaway[7], Jeffrey L. Neul[8], Alana Salvucci[9], Rachel McMinn[9], Julie Jordan[9]

[1]The Kids Research Institute Australia, Centre for Child Health Research, The University of Western Australia, Perth, WA, Australia; [2]Departments of Pediatrics & Neurology, Texas Children's Hospital, Baylor College of Medicine, Houston, TX, USA; [3]Department of Pediatrics-Neurology, Children's Hospital of Colorado, Anschutz Medical Campus, University of Colorado School of Medicine, Aurora, CO, USA; [4]Department of Neurology, Boston Children's Hospital, Harvard Medical School, Boston, MA, USA; [5]Departments of Pediatrics, Neurological Sciences, and Biochemistry, Rush University Medical Center, Chicago, IL, USA; [6]Isabelle Rapin Division of Child Neurology, Montefiore Medical Center, Albert Einstein College of Medicine, New York, NY, USA; [7]Sydney Children's Hospital Network, The University of Sydney, Sydney, Australia; [8]Department of Pediatrics-Neurology, Monroe Carell Jr. Children's Hospital at Vanderbilt, Vanderbilt University Medical Center, Nashville, TN, USA; [9]Neurogene Inc., New York, NY, USA.

medicalinfo@neurogene.com

© 2026 Neurogene Inc.
All rights reserved.

INTRODUCTION

- Rett syndrome (RTT) is a rare neurodevelopmental disorder caused by loss-of-function variants in the *MECP2* gene encoding MeCP2, a protein critical for normal function in the brain and nervous system.[1,2]
 - Hallmark features include developmental regression, impaired gross motor function, and limited acquisition of complex motor skills.[1–4]
- Caregivers report that even modest gains in motor function can enhance patient autonomy, reduce complications, and ease caregiving burden.[3]
- NGN-401 is an investigational AAV9 gene therapy that delivers the full-length human *MECP2* transgene, incorporating EXACT™ transgene regulation technology designed to enable controlled MeCP2 expression on a cell-by-cell basis, and is administered intracerebroventricularly for the treatment of RTT.[5]
- The data presented here show improvement in gross motor function in participants who received NGN-401 in the Phase 1/2 trial.

METHODS

- The Phase 1/2 open-label trial (NCT05898620) in females with classic RTT, age ≥4, with a baseline Clinical Global Impression – Severity (CGI-S) score of 4–6, is evaluating the safety, tolerability, and efficacy of NGN-401.[6]
- 10 participants received 1E15 vg NGN-401 (age 4–10 cohort, n=8; age ≥11 cohort, n=2).
- The data cutoff was June 16, 2026, yielding a minimum of 12 months and up to 30 months of follow-up across the 10 participants.
- Gross motor function was assessed in 2 ways:
 - Through acquisition of developmental milestones based on the criteria in the Embolden registrational trial evaluating NGN-401.[7]
 - The Rett Syndrome Gross Motor Scale (RSGMS),[8] a validated clinical tool assessing 15 gross motor tasks for RTT (**Figure 1**). Tasks are grouped into three subdomains: sitting, standing/walking, and challenge skills. A higher RSGMS score indicates better gross motor function.

Figure 1. The RSGMS assesses gross motor abilities.

Assessing 15 key gross motor abilities
Evaluates movements ranging from sitting and standing to walking and "challenge" skills.

Higher scores indicate better function
A higher total score reflects ability to perform tasks with less physical assistance.

Increase in 3+ points signals meaningful improvement
An increase of 3+ points is considered a meaningful gain in motor ability.

Each of the 15 items is scored on a scale of 0–3, for a possible total score of 45

Score	0	1	2	3
Description	Maximal assistance*	Moderate assistance	Minimal assistance	No assistance

*Item is also scored 0 if a participant is unable to perform.

RSGMS analysis

- RSGMS testing was videotaped in clinic, and scores were assessed by central independent raters.
- Replicable and best attained scores were assessed relative to baseline, to account for variability across visits (e.g., fatigue, illness, attention, apraxia) and align with established responder frameworks in rare neurological diseases.[9,10]
- Reproducibility of best score was confirmed via ≥2 documented central ratings and/or centrally reviewed videos performed outside the assessment. Repeat confirmation was not required when the best score occurred at the most recent assessment as it was deemed an emergent skill.

Table 1. Phase 1/2 baseline participant demographics.

Characteristic	All participants (N=10)
Age at dosing, years, range	4–18
Baseline CGI-S score*	4–6
Genetic variant severity	Mild to severe
Time post-dosing	12–30 months
Baseline ambulatory status	Ambulatory: 60%

*Score of 4 indicates 'Moderately ill'; score of 5 indicates 'Markedly ill'; score of 6 indicates 'Severely ill".
CGI-S, Clinical Global Impression – Severity.

RESULTS

- **Mean RSGMS score increased by 6 points**, from 20.5 (range 9–37) at baseline to 26.5 (range 13–42) post treatment.
- When normalized to a 1-year rate, NGN-401 treatment resulted in statistically significant gain vs. a decline in natural history (**Figure 2A**).
- Ambulatory participants had a mean **8.3-point increase** in post-treatment total scores; non-ambulatory participants had a **2.5-point increase**.
- Skill level improvement (**Figure 2B** and **2C**) was assessed in eligible participants—those without full independence in that skill at baseline.
- Ambulatory participants demonstrated improvement across RSGMS fundamental and complex motor tasks (**Figure 2B**), while non-ambulatory participants had gains primarily in seated balance, standing duration, and sit to stand (**Figure 2C**).
- **All participants** achieved independent sitting following treatment (**Figure 2B** and **2C**).

Figure 2. RSGMS score improvements.

Mean RSGMS total-score gain exceeded natural history normalized to 1-year rate

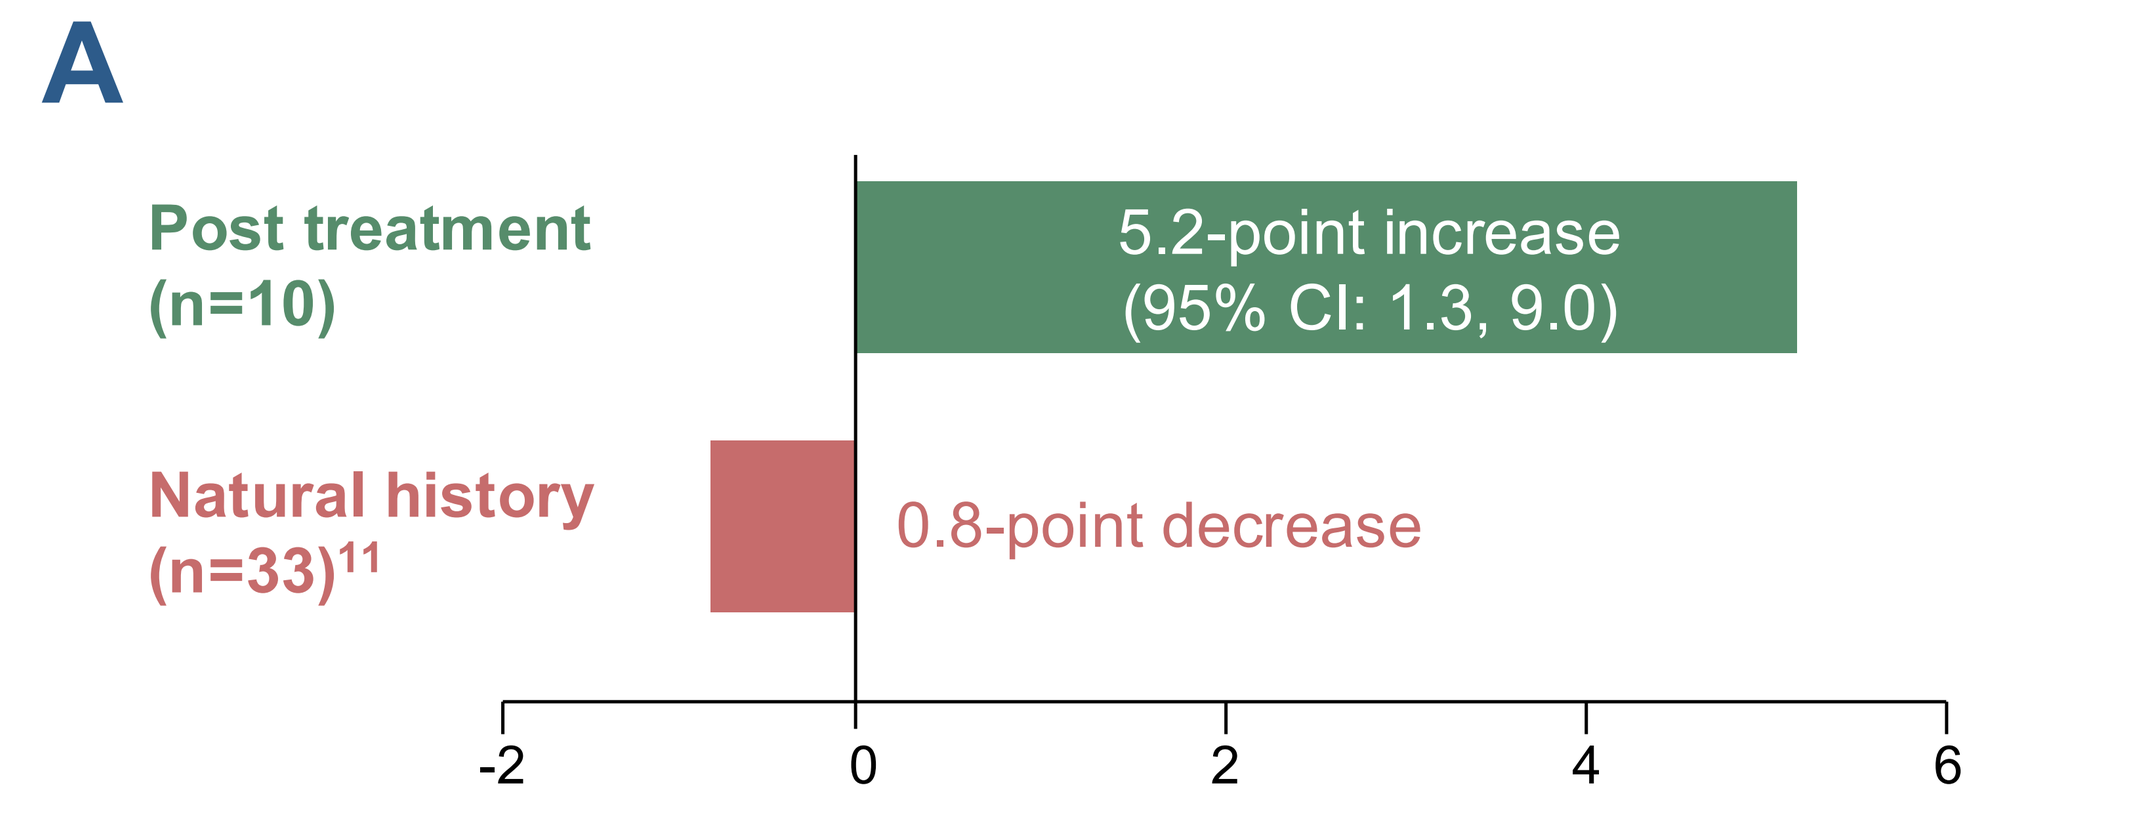
A
Post treatment (n=10)
5.2-point increase (95% CI: 1.3, 9.0)
Natural history (n=33)11
0.8-point decrease
-2
0
2
4
6

P<0.001. One sample Wilcoxon test.
CI, confidence interval.

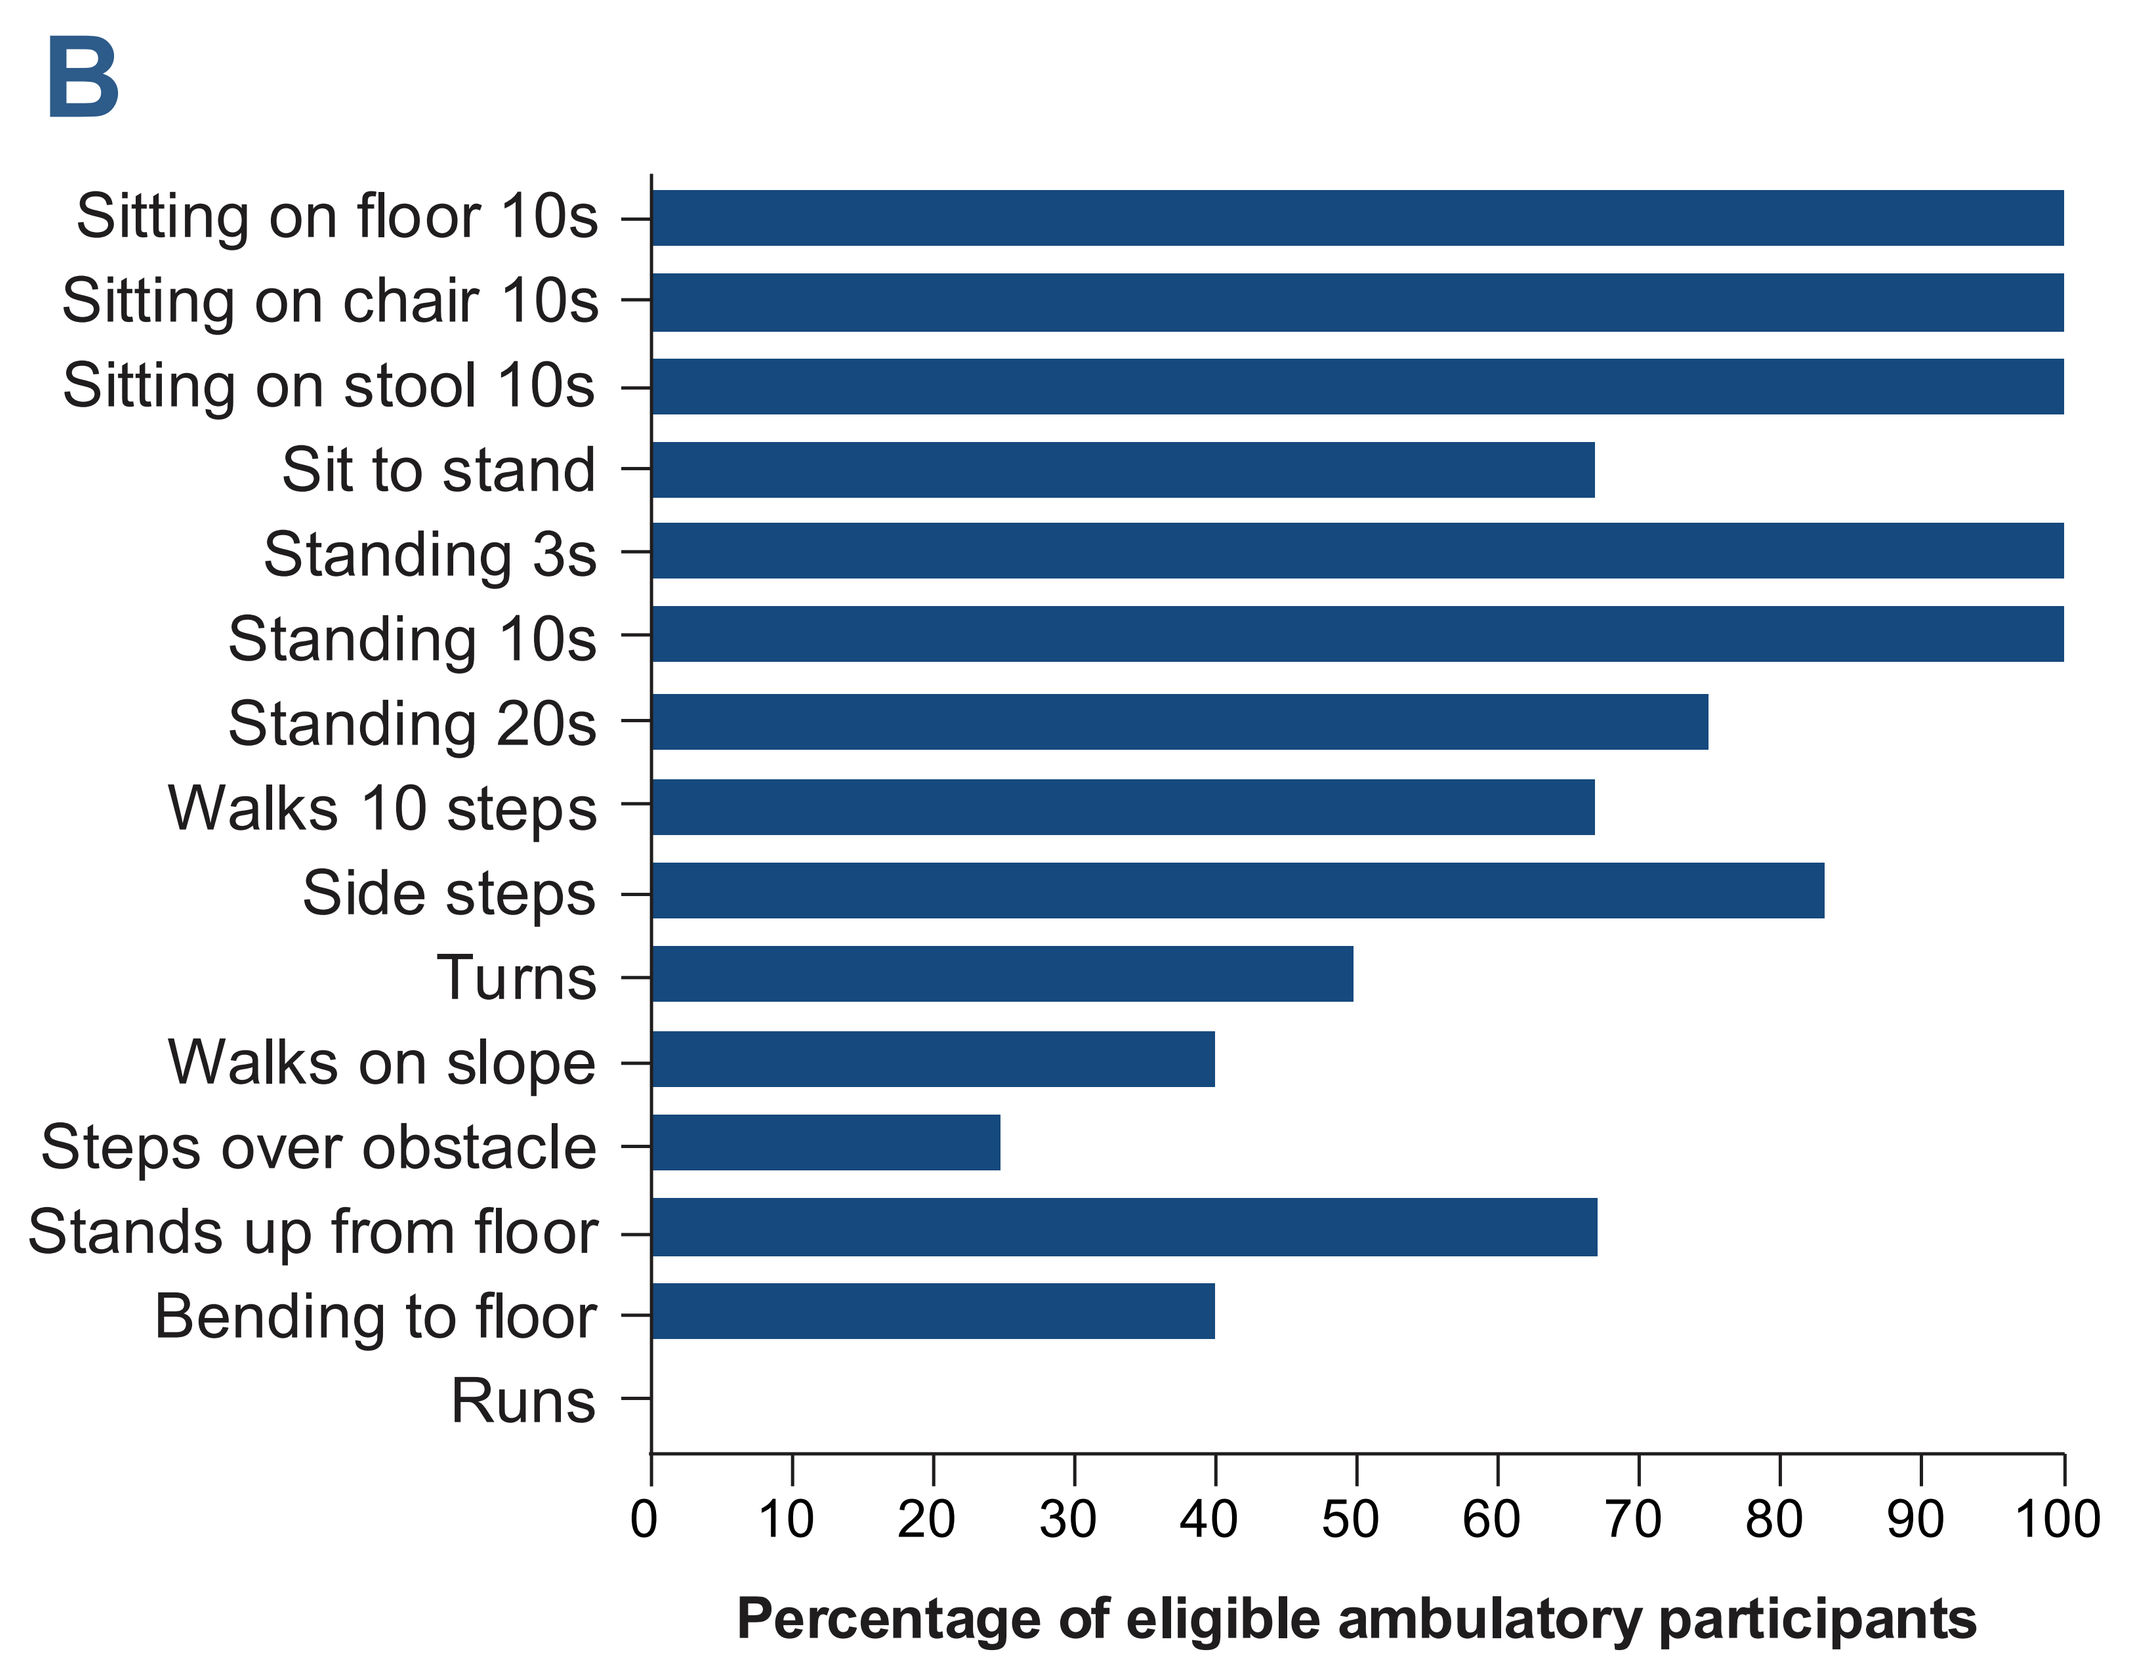
B
Sitting on floor 10s
Sitting on chair 10s
Sitting on stool 10s
Sit to stand
Standing 3s
Standing 10s
Standing 20s
Walks 10 steps
Side steps
Turns
Walks on slope
Steps over obstacle
Stands up from floor
Bending to floor
Runs
0 10 20 30 40 50 60 70 80 90 100
Percentage of eligible ambulatory participants

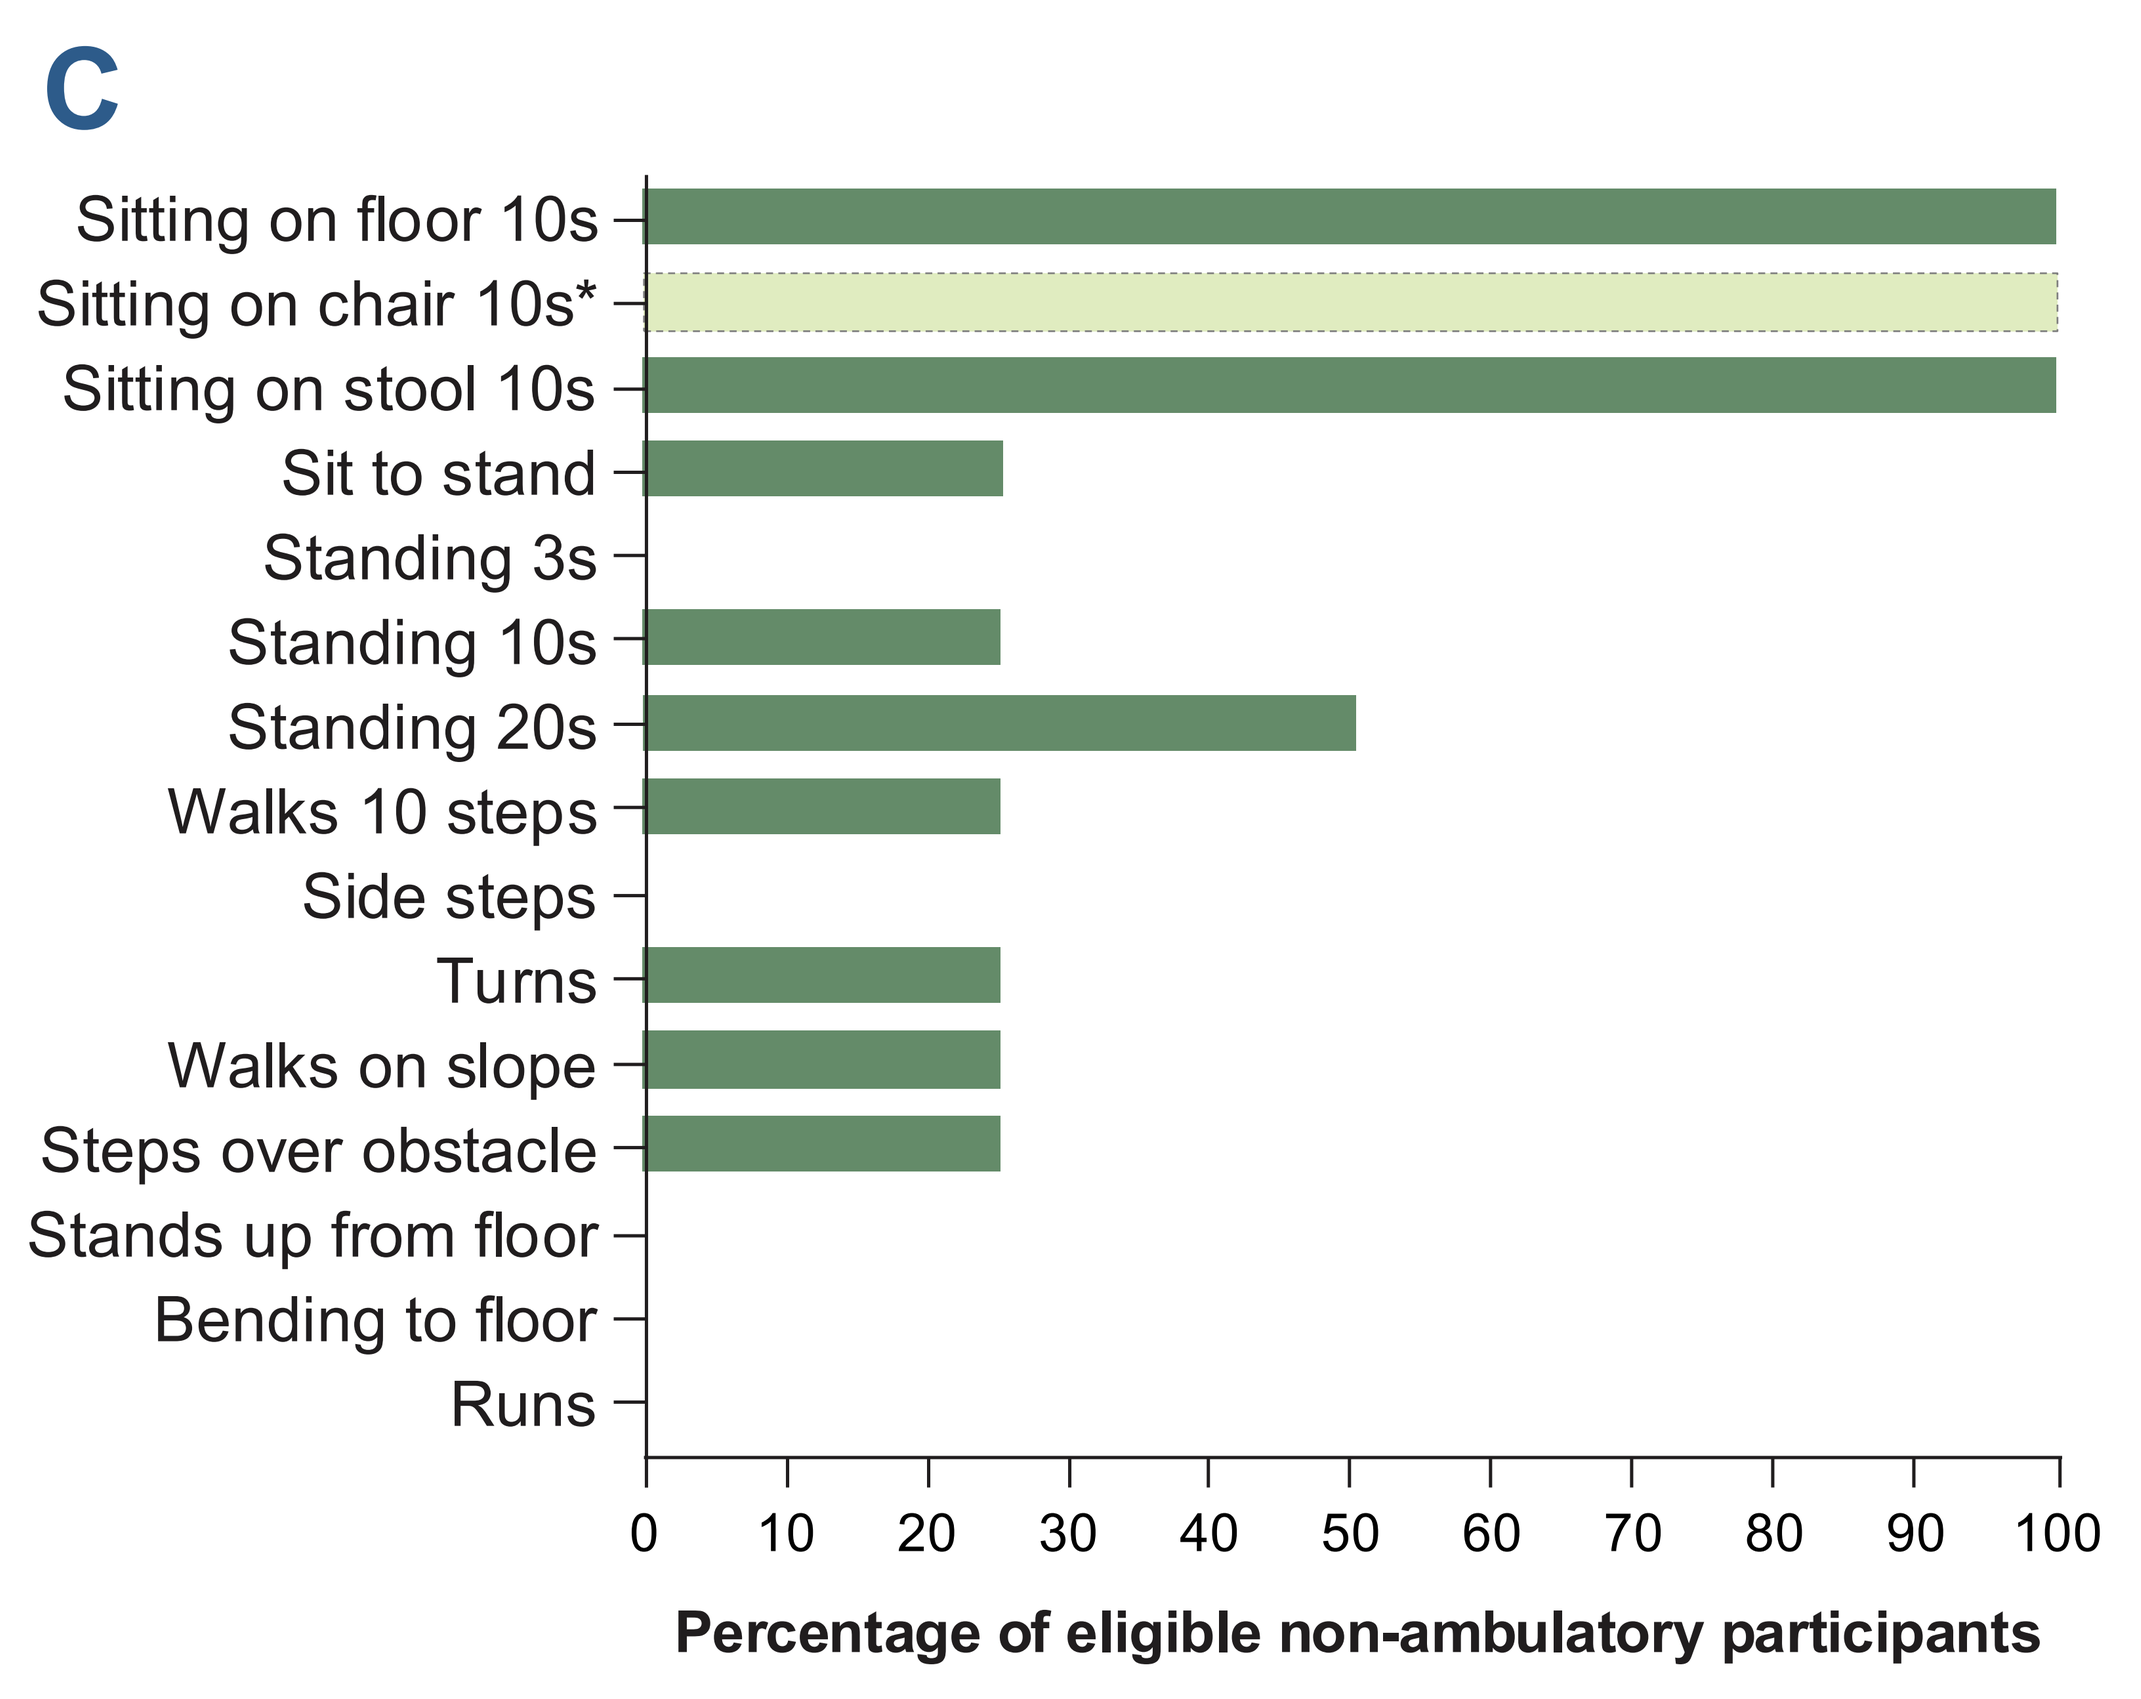
C
Sitting on floor 10s
Sitting on chair 10s*
Sitting on stool 10s
Sit to stand
Standing 3s
Standing 10s
Standing 20s
Walks 10 steps
Side steps
Turns
Walks on slope
Steps over obstacle
Stands up from floor
Bending to floor
Runs
0 10 20 30 40 50 60 70 80 90 100
Percentage of eligible non-ambulatory participants

*All participants were able to complete this task with no assistance at baseline.
A gain is defined as a 1-point or more increase in the item from baseline to any visit. Eligible participants are defined as those who could not perform with full independence at baseline.
RSGMS, Rett Syndrome Gross Motor Scale; s, seconds.

Transition skill gains

5/7 improved

SIT TO STAND: Gains reduce physical burden on caregiver and enhance independence

n=7 could not independently perform at baseline.

Pt:3: Sit to stand with less assistance, RSGMS score=1

BASELINE: Non-ambulatory, required maximum support (RSGMS score=0) to transition from sitting to standing and the support of 2 people to maintain standing.

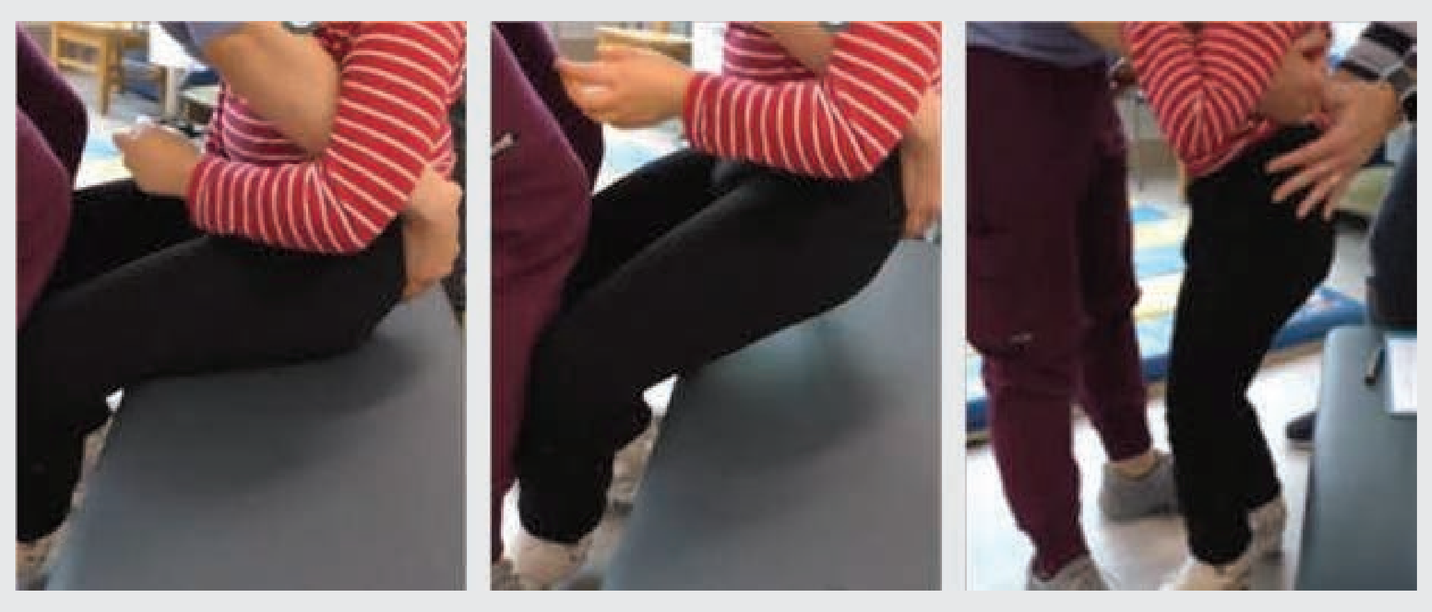

POST TREATMENT: Improved functional mobility, requiring only moderate assistance (RSGMS score=1) for sit-to-stand transfers and the support from a single individual to maintain standing.

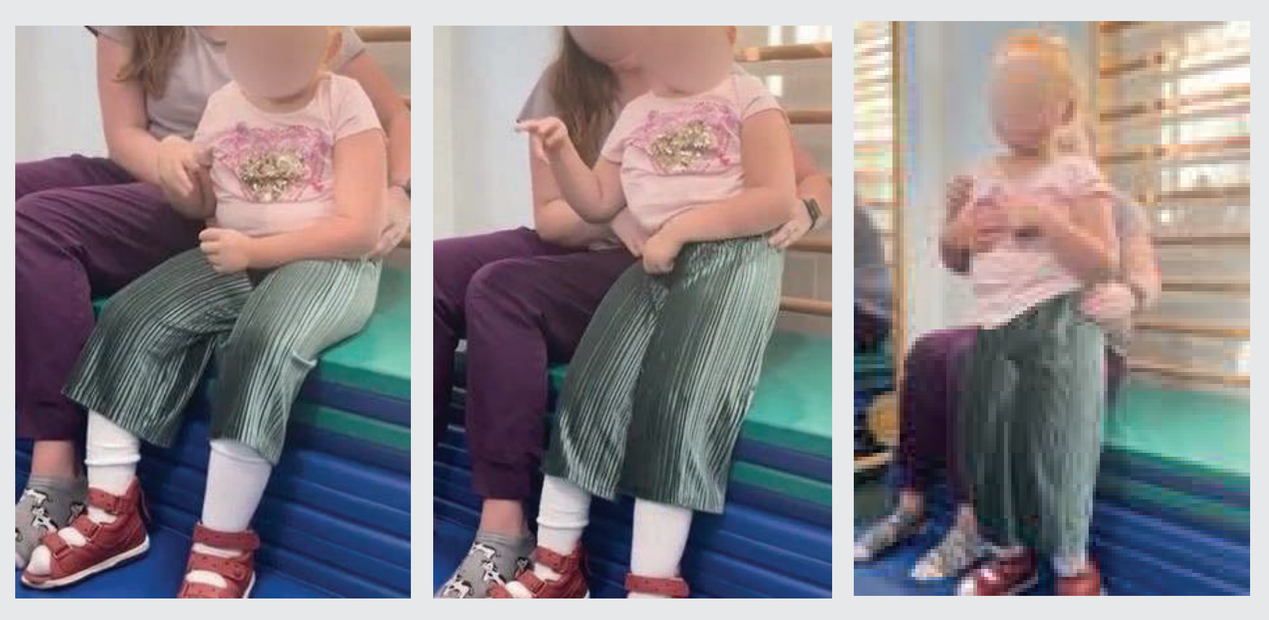

STANDS UP FROM FLOOR:
Complex motor planning allows for greater autonomy and reduced caregiver physical burden

5/6 improved

n=6 ambulatory participants could not independently perform at baseline.

Pt:8: Standing up from floor independently, RSGMS score=3

BASELINE: Ambulatory, needed moderate assistance to stand up from floor, RSGMS score=1

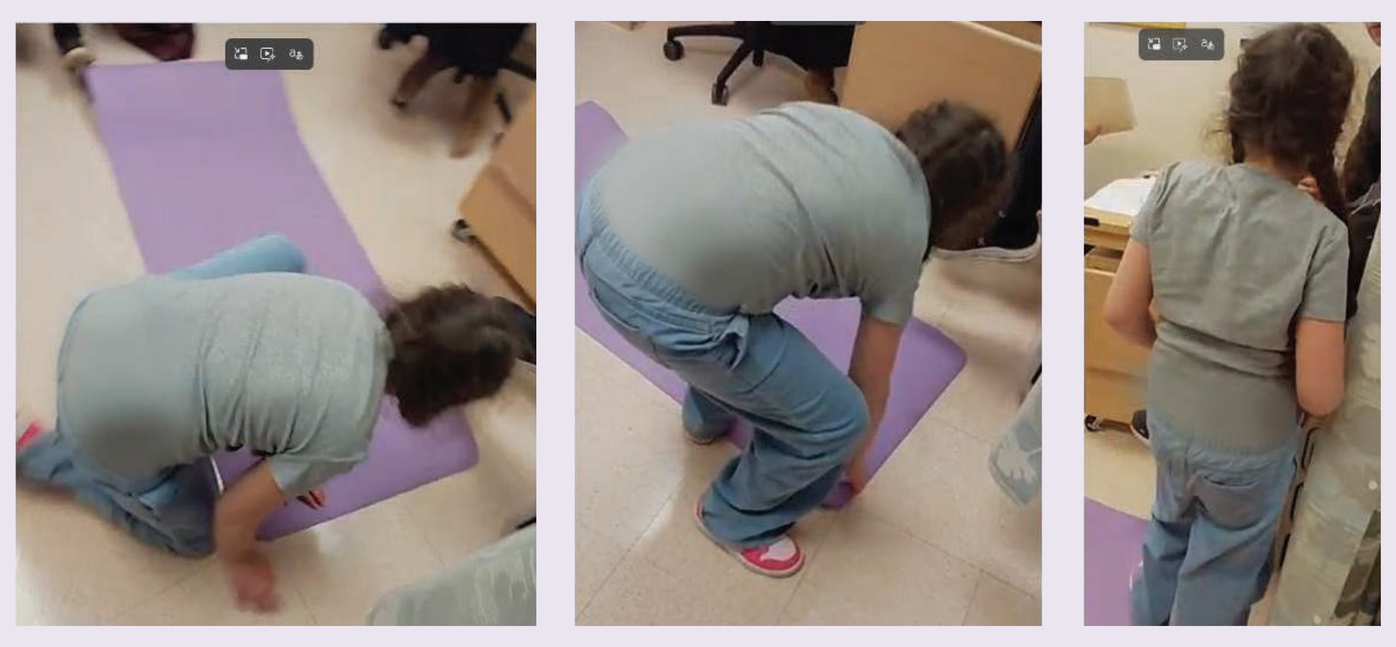

Pt:8 New milestone post treatment

BENDING TO FLOOR:
Enables picking up toys on own and exploring the world at ground level without help

2/5 achieved full independence

n=5 ambulatory participants could not independently perform at baseline.

Pt:2: Bends to floor and recovers to standing independently, RSGMS score=3

BASELINE: Ambulatory, unable to bend to floor and recover to standing independently, RSGMS score=0

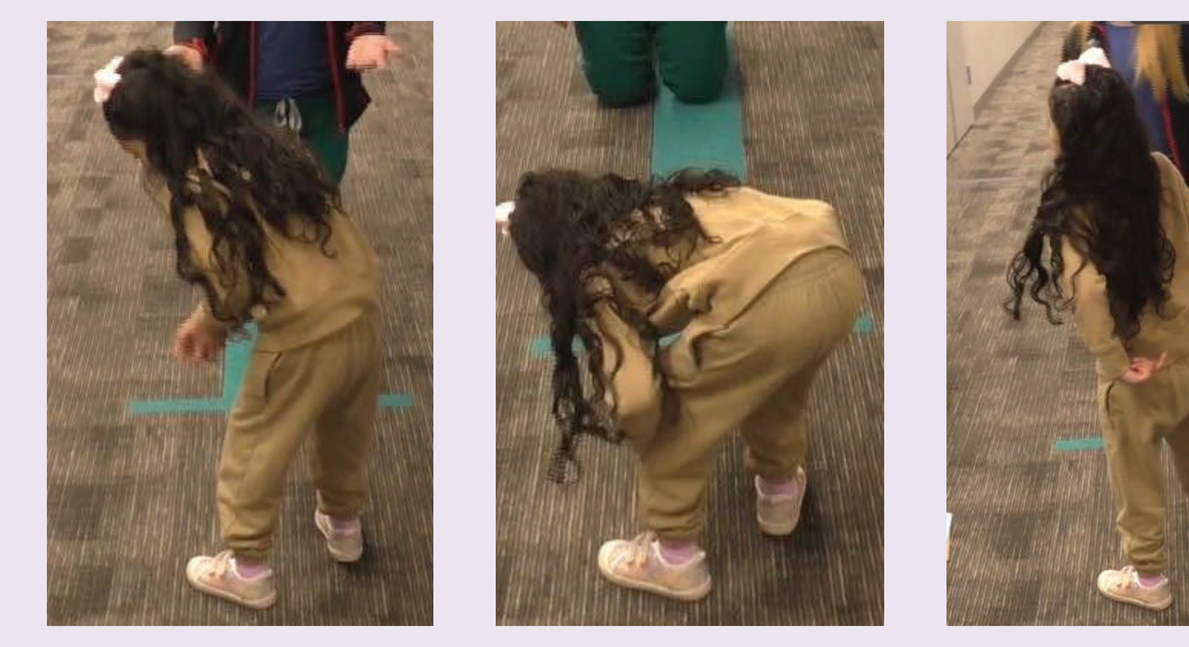

Pt:2 New milestone post treatment

Real-world impact

Real-world impact of improved gross motor function translates to increased participation in family routines

Pt:4: Baseline: Non-ambulatory, unable to transition from supine to sitting independently

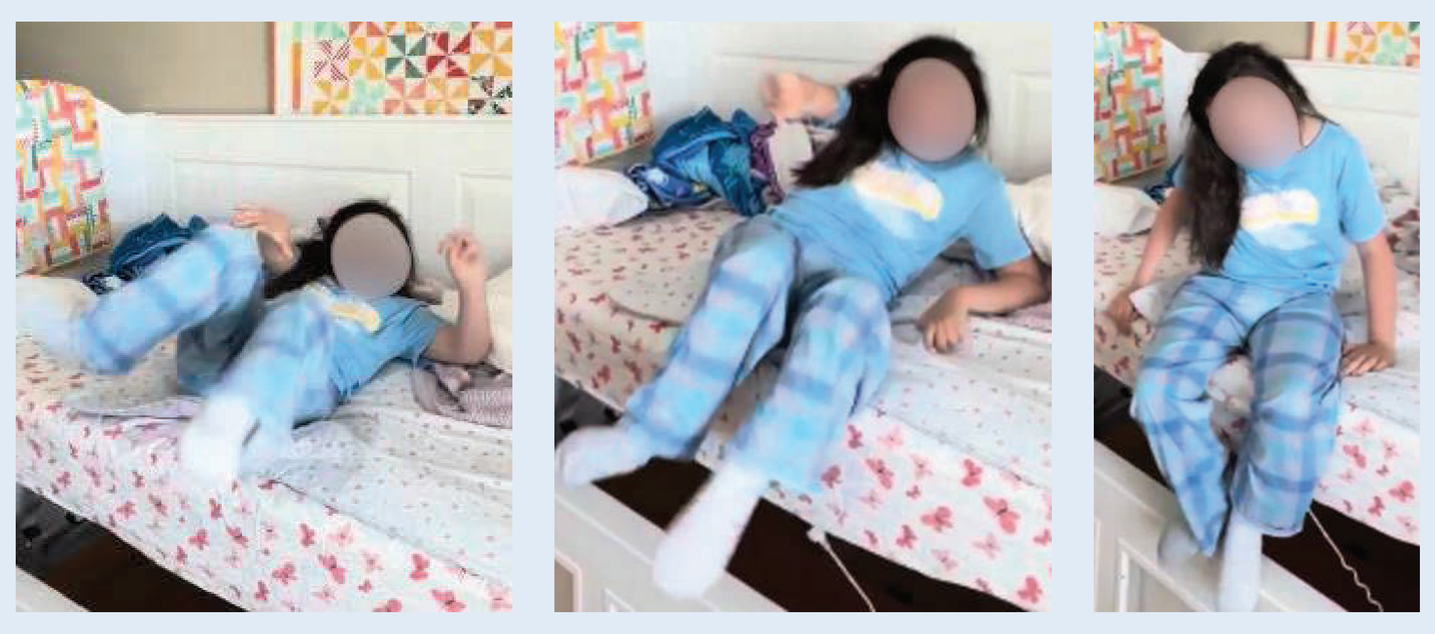

Pt:4 New milestone post treatment

Sitting up in bed independently:
Self-initiated transitioning from supine to sitting up position on caregiver request; demonstrating motor planning, self-initiated routine, improved postural control, increased autonomy, and increased engagement with her surroundings.

Pt:7: Baseline: Ambulatory, unable to transition from prone position on floor to sitting independently

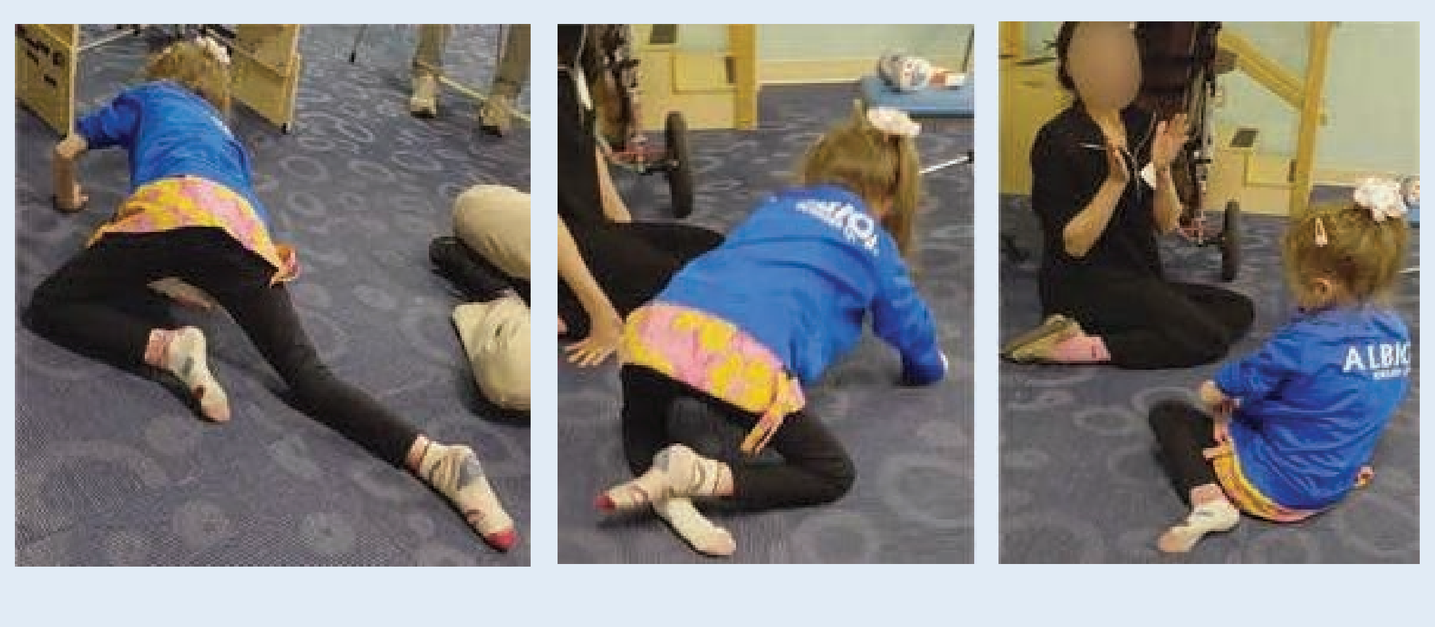

Pt:7 New milestone post treatment

Transition from lying to sitting independently:
Self-initiated repositioning from the floor into sitting as caregiver applauds, demonstrating improved motor planning, postural control, increased autonomy, and increased engagement with her surroundings.

Pt:1: Baseline: Ambulatory, unable to ascend/descend stairs independently, impaired hand function, dropped items quickly

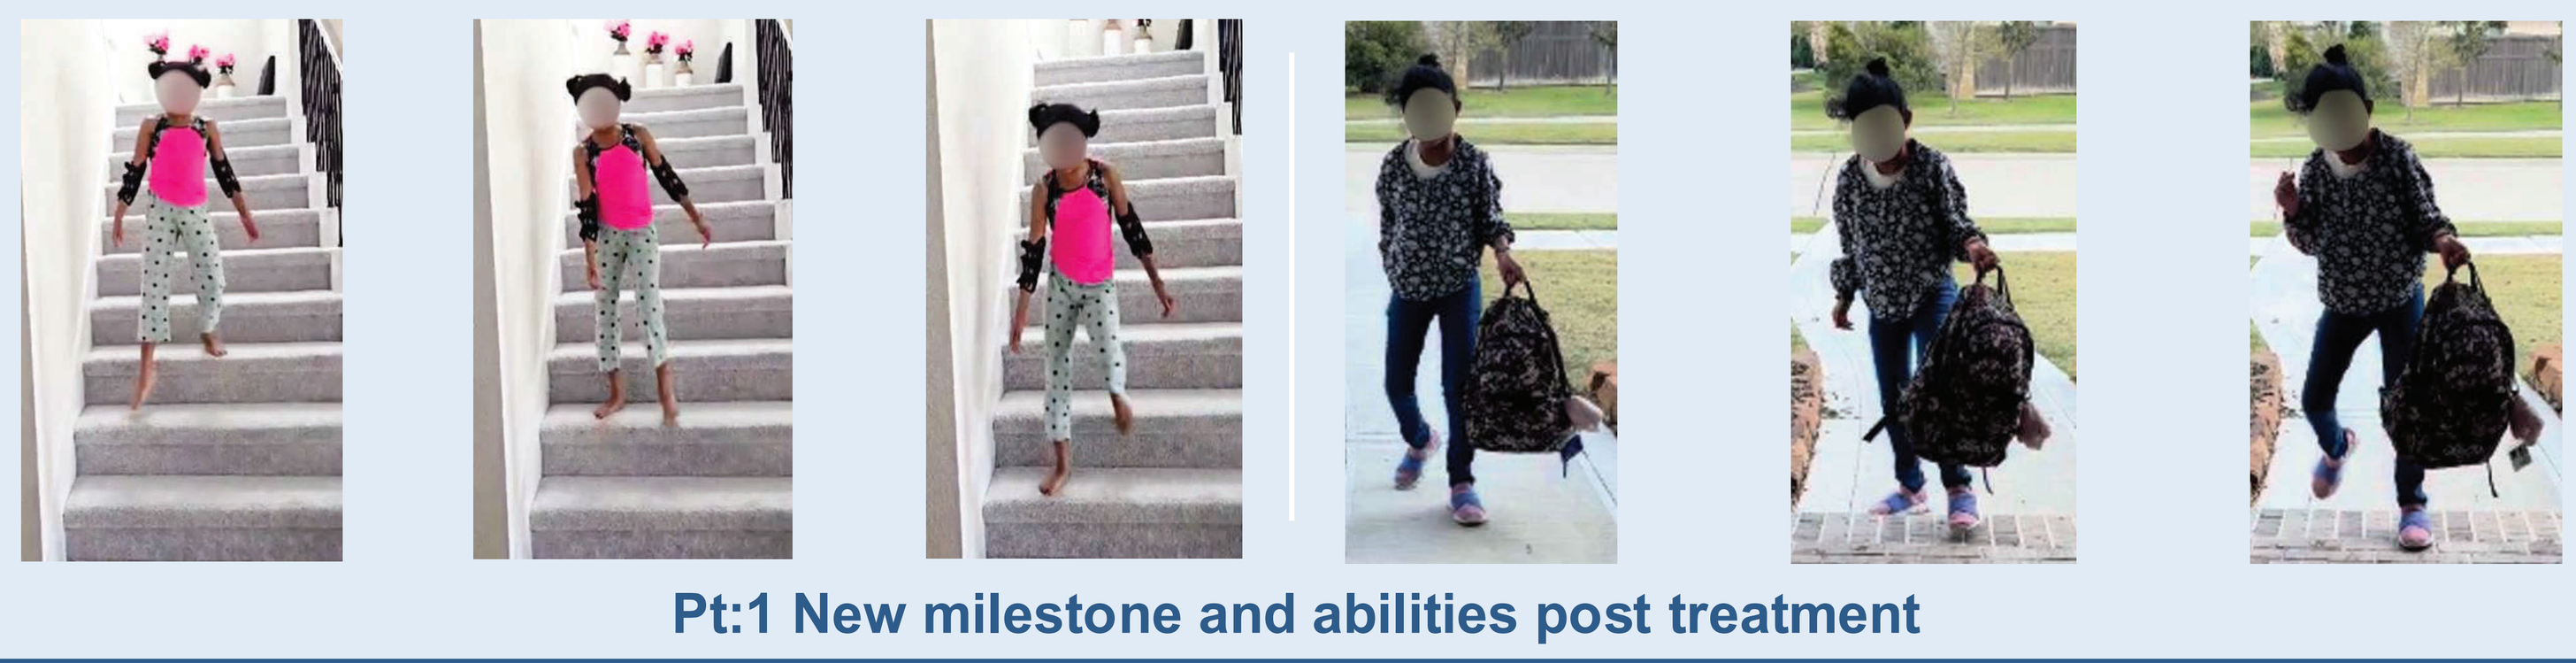

Pt:1 New milestone and abilities post treatment

Descending stairs independently:
Autonomous navigation of stairs with appropriate motor planning, balance, and postural control as caregiver waits at the bottom, demonstrating safe and independent home mobility.

Coming home from school:
Able to navigate walkway and stairs up to house while carrying school backpack; meaningful routine requiring complex motor planning, balance, grip strength, and functional independence.

Pt, participant.

KEY CONCLUSIONS

1. Treatment with NGN-401 resulted in a **mean RSGMS increase of 6 points**, representing a clinically meaningful improvement on a scale that declines over time in Rett syndrome natural history.[11–13]
2. **Spanning across pediatric and adult participants, skill gains followed a hierarchical pattern** tied to baseline status: **More advanced skills** in ambulatory participants and **core postural control improvements** in non-ambulatory participants.
3. Both groups showed transition skill gains, pointing to **enhanced motor planning, coordination, and execution, abilities typically impaired in RTT**.
4. RSGMS gains translated into **new functional skills, developmental milestone acquisition, and greater daily independence**, consistent with caregiver-defined clinically meaningful outcomes and a potential reduction in caregiver burden.[14]

References

1. Neul JL *et al. Ann Neurol* 2010; 68 (6): 944–950. **2.** Einspieler C *et al. Neurosci Biobehav Rev* 2019; 98: 320–332. **3.** Neul JL *et al. J Neurodev Disord* 2014; 6 (1): 20. **4.** May D *et al. BMC Neurol* 2023; 23 (1): 141. **5.** Ross PD *et al. Sci Transl Med* 2025; 17 (792): eadq3614. **6.** ClinicalTrials.gov NCT05898620. Available at: https://clinicaltrials.gov/study/NCT05898620. Accessed September 2026. **7.** Neul JL *et al.* Poster 45 presented at IRSF 2026; Prior Lake, MN, USA, 29 June – 1 July 2026. **8.** Downs J *et al. PLoS One* 2016: 11 (1): e0147555. **9.** Strauss KA *et al. Nat Med* 2022; 28 (7): 1390–1397. **10.** Finkel RS *et al. N Engl J Med* 2017; 377: 1723–1732. **11.** Bisgaard AM *et al. Front Neurol* 2026; 16: 1702703. **12.** Bisgaard AM *et al. Am J Med Genet A* 2021; 185 (12): 3683–3693. **13.** Bisgaard AM *et al.* Poster presented at ASCEND 2025; Boston, MA, USA, 9–11 June 2025. **14.** International Rett Syndrome Foundation (IRSF), Rett Syndrome Research Trust (RSRT) – Voice of the Patient Report: Living with Rett Syndrome. Available at: https://rettpfdd.org/site/assets/files/1/2022-rett-syndrome-voice-of-the-patient-report.pdf. Accessed September 2026.

Acknowledgments and Funding

The authors would like to extend gratitude to the participants, families, and site staff who were involved in the Phase 1/2 trial. Special thanks to the whole Neurogene team that supported this work. Medical writing and editorial support were provided by Nicole L. Day, PhD, MWC® of Porterhouse Medical.

This trial and medical writing support were funded by Neurogene Inc.

Presented at the 55th Child Neurology Society Annual Meeting, October 14–17, 2026, Montréal, Québec, Canada